Stability and comparability of an amorphous drug prepared by different spray drying processes: atomic Pair-wise distribution functions (PDF) using conventional X-ray diffraction versus high energy synchrotron radiation

Hector Novoa de Armas[1], Marcus Brewster[1], Detlef Beckers[2], Milen Gateshki[2], Chris Benmore[3], Stephen Byrn[4]

[1] Pharmaceutical and Material Sciences, Johnson & Johnson Pharmaceutical Research & Development, a division of Janssen Pharmaceutica NV, Turnhoutseweg 30, 2340 Beerse, Belgium

[2] PANalytical B.V. Lelyweg 1 (7602 EA) PO Box 13, 7600 AA, Almelo, The Netherlands

[3] Argonne National Laboratory, 9700 S. Cass Ave, Argonne, IL 60439, USA

[4] Department of Industrial and Physical Pharmacy, Purdue University, 575 Stadium Mall Drive, West Lafayette, IN 47906, USA

INTRODUCTION

The amorphous state is of significant interest within the pharmaceutical industry, particularly as a possible means to enhance aqueous solubility of APIs. An important practical barrier to the development of amorphous APIs in product development is the lack of reliable methods for fingerprinting. The Atomic Pair distribution function (PDF) methods have been suggested as an alternative approach for fingerprinting amorphous[1]. The PDF technique utilizes a Fourier transformation of the X-ray powder diffraction (XRPD) data to produce a trace in a coordinate system. The y-axis in the PDF trace is corresponding to the probability of finding two atoms separated by a distance stipulated by the x-axis[2]. The PDF hence assesses the inter-atomic distances of the material. The accuracy of this assessment of inter-atomic distances is directly proportional to the energy of the utilized radiation source.

PURPOSE

To evaluate the relative merits of an atomic Pair-wise distribution function (PDF) generated using conventional XRPD (Mo or Ag-sourced) and synchrotron radiation in assessing process variations in amorphous drug preparation by spray drying.

METHODS

Formulations of the same API were prepared by spray drying having different solvent composition. PDF analyses of these spray dried powders (SDPs) were conducted using conventional laboratory XRPD instrumentation and compared to those obtained using high energy X-ray synchrotron data to measure diffuse scattering intensities which contains information related to local ordering in the sample.

PDF analyses conventional XRPD:

Total scattering measurements were performed on a PANalytical X'Pert PRO MPD multipurpose diffractometer with X'Celerator detector. The X-ray tubes were operated at 60 kV (K-α radiation: λ=0.7107 Å (Mo-radiation), λ=0.5609 Å (Ag-radiation)). Samples were loaded in glass capillaries (2 mm external diameter). The data was collected in the angular 2θ interval 3 to 150°, which corresponds to a Q_{max} value of 17.1 $Å^{-1}$ (Mo) or 21.6 $Å^{-1}$ (Ag). An optimized variable counting time strategy was adopted to counteract the decrease of the scattered intensity at the highest angles due to the X-ray form factor. The total data collection time for each sample was about 22 hours. The data was processed using the software RAD[4].

PDF analyses synchrotron XRPD:

The synchrotron data was collected to high scattering vector Q (Q > 20 $Å^{-1}$) at the Advanced Proton Source at Argonne National Laboratory on beamline 11-ID-B (beam size 1mmx1mm). The powder samples were packed in capton capillaries of 1.1mm outside diameter and were analyzed at 60 keV (0.2127 Å). Two packing of each of the four samples was prepared and data was collected for a total of 33 min. The data was collected using an area detector and processed with the PDFgetx2 program[5].

RESULTS

PDF analyses conventional XRPD:

1. Experiment using Mo radiation:

The final PDF traces of the SDP formulations (samples A-D) prepared by different solvent composition are shown below (for better visibility of the data at small radial distances the data is plotted only to Q = 10 Å):

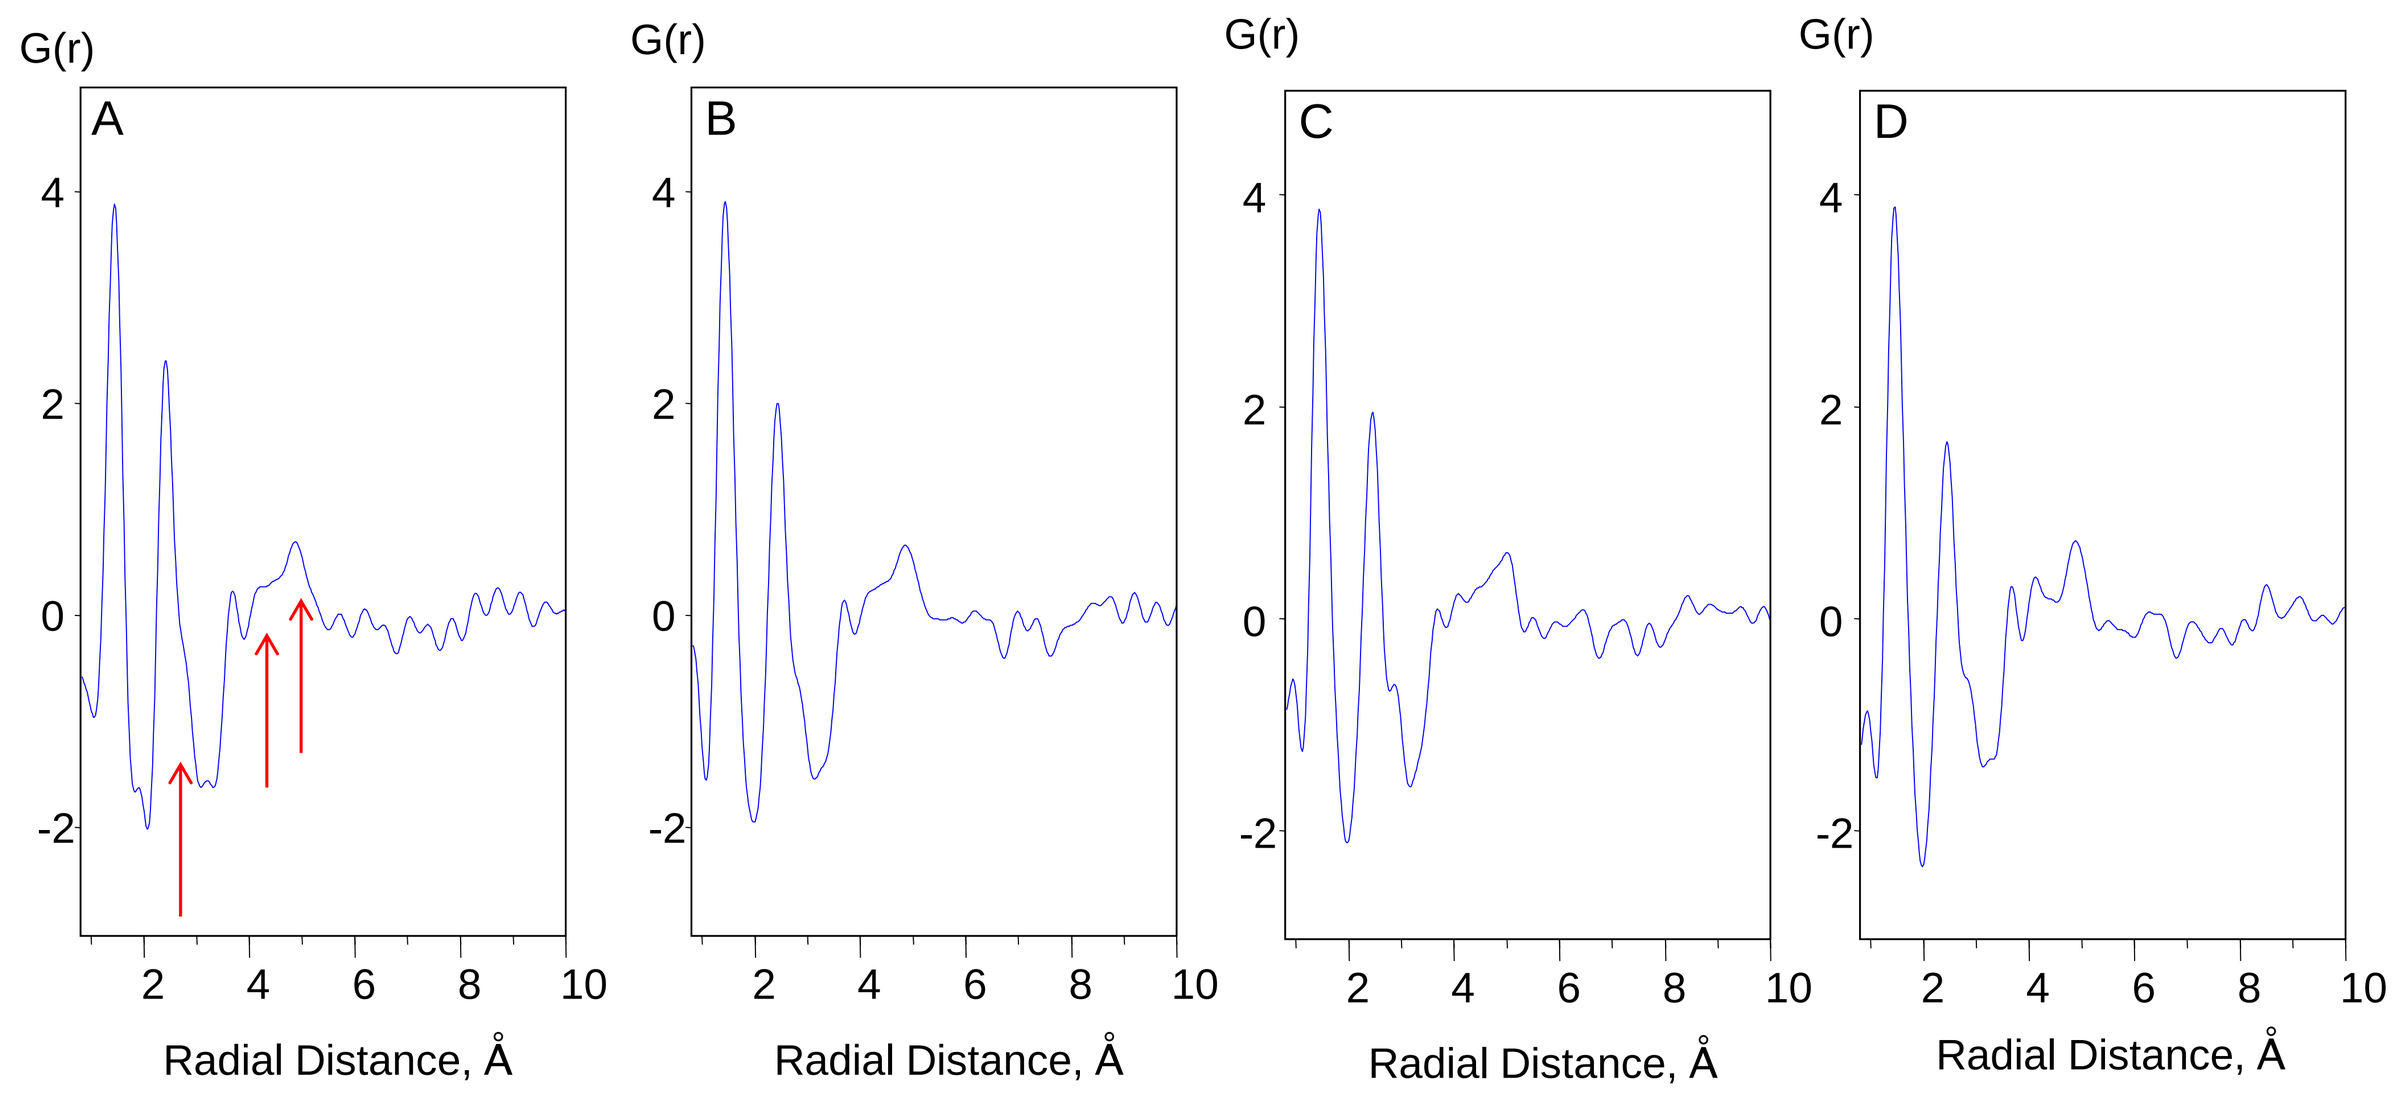
G(r)
A
B
C
D
Radial Distance, Å

The red arrows indicate the regions with the main variations between the PDF traces. The average PDF pattern G(r) and the difference between the average PDF pattern and the PDF pattern of each sample are plotted below:

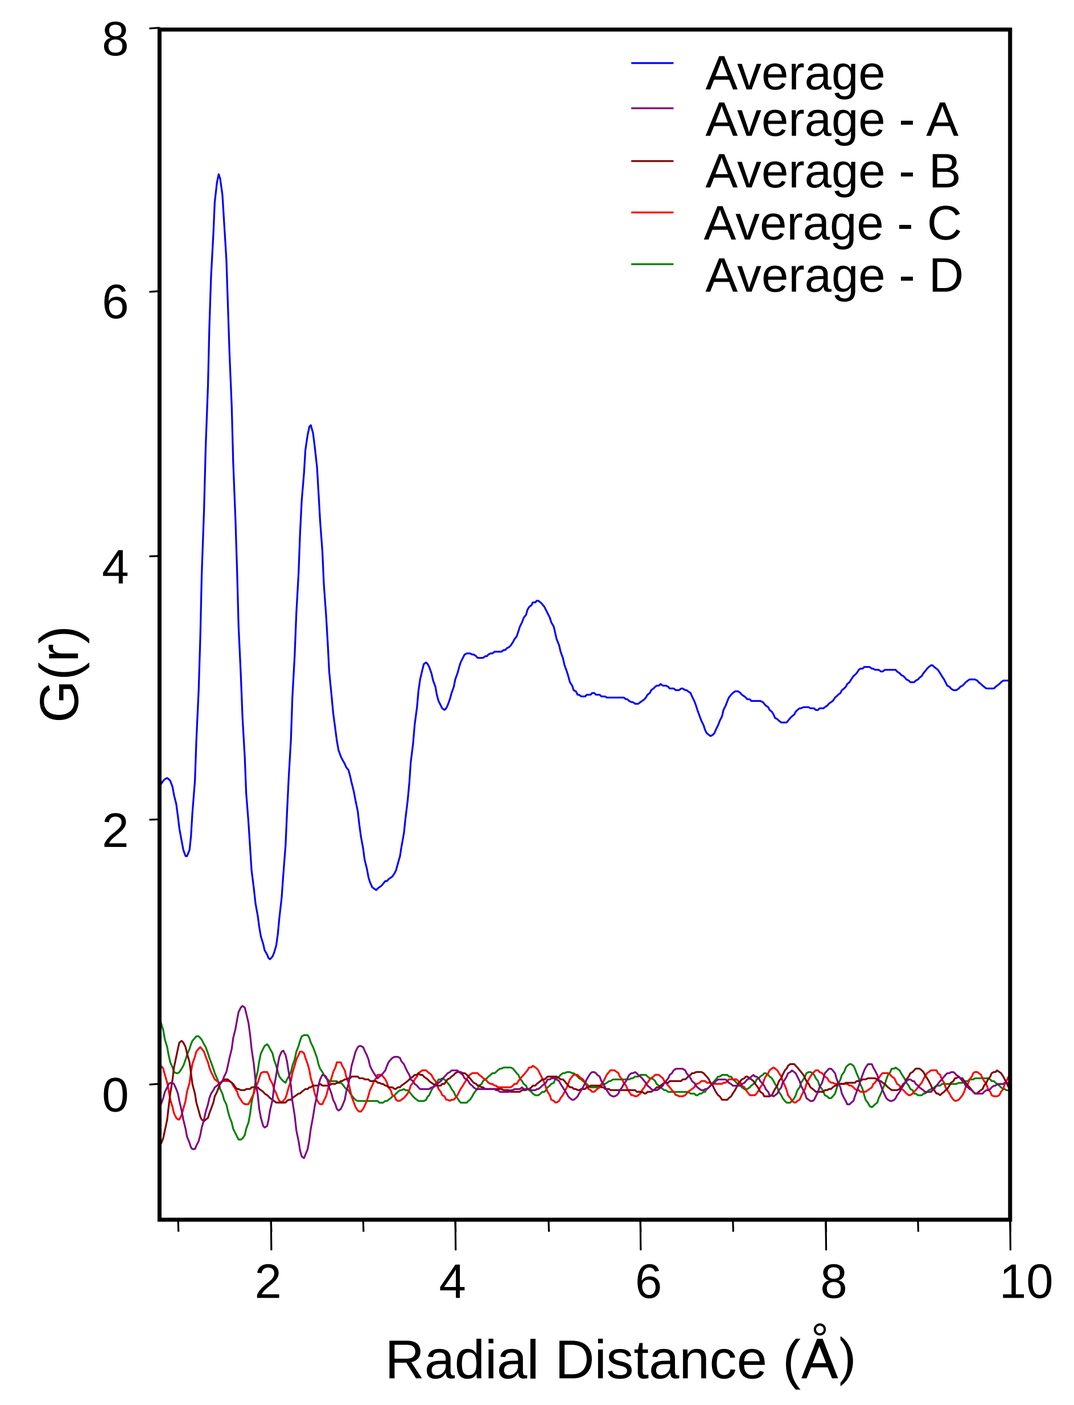
Average
Average - A
Average - B
Average - C
Average - D
G(r)
Radial Distance (Å)

The observed differences are relatively small . To clarify whether these variations are sample related or artifacts from the data treatment and/or counting artifacts, additional measurement with higher energy radiation (Ag) were performed on samples A and D. The observed small features in the Mo-radiation data were not clearly reproduced with Ag radiation and do not seem to be determined by the sample structure. The observed fluctuations in the PDFs are not significant enough to be interpreted as sample property.

PDF analyses synchrotron XRPD:

The Figure below shows the average PDF for the different SDP formulations. This is also representative of each individual sample since the data from each sample are similar. Note that the magnitude of G (r) is about 5.

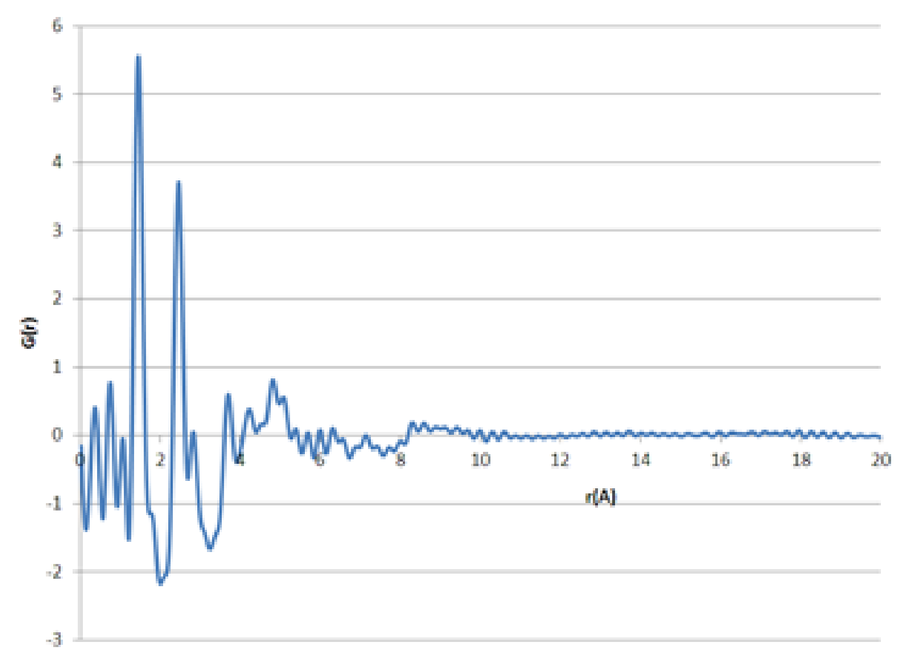

In order to gain insight into variations in the X-ray source, the PDF plots (G(r) vs. r(Å)) from the various data sets were compared to each other. The G(r) from each of the ten 200 sec runs was averaged for each r(Å) value. Then the G(r) value from each of the 10 runs was subtracted from the average value to give Delta G(r).
The largest difference seen was about 0.346 and the average of the largest deviations is 0.176:

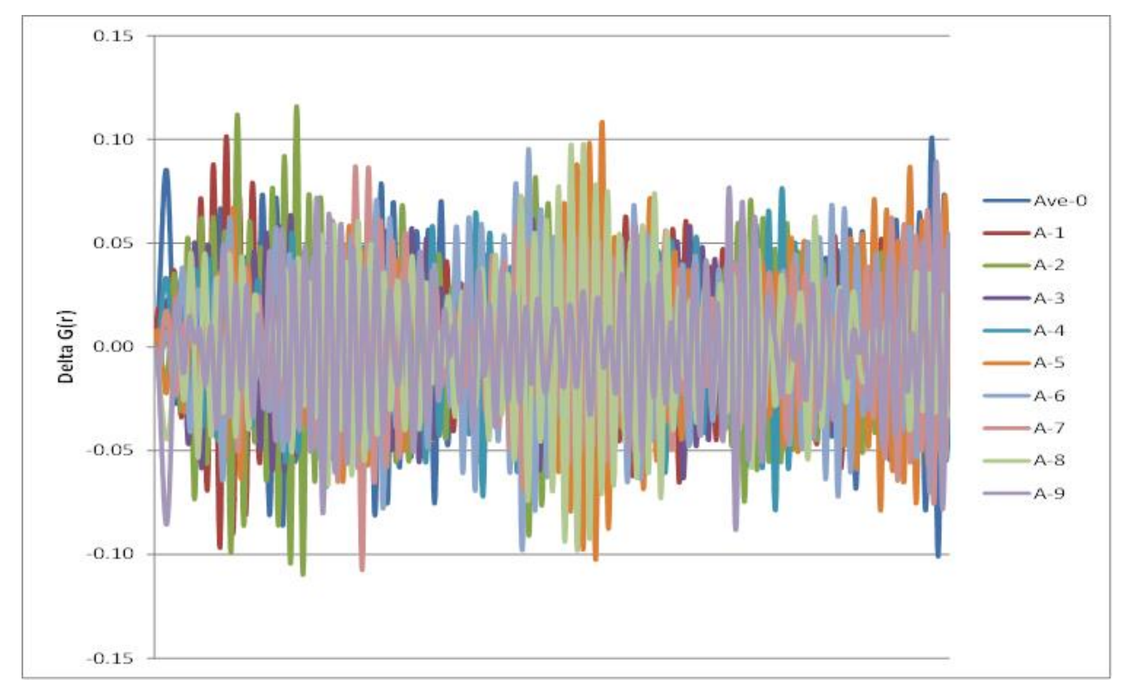
Delta G(r)
Ave-0
A-1
A-2
A-3
A-4
A-5
A-6
A-7
A-8
A-9

Since the magnitude of G(r) was about 5, the Delta G(r) is less than 5%. This result shows that the data are reproducible.

In order to determine the effect of sample packing the average PDF pattern (G(r) vs. r(Å)) for each of the two sample packing was calculated and the difference between the G(r) was then determined:

Sample	Largest Difference between the average G (r) and for the individual packing
A	0.230
B	0.198
C	0.261
D	0.225
Average	0.229

The average deviation is slightly larger than for the repeated runs but still shows very good agreement between the packings. This gives confidence that the results are not dependent on how the capillary was filled or other factors related to subsampling for the capillary.
Besides, the average PDF for all four samples was compared to the PDF of each individual sample to attempt to discern any differences in structure. The differences are quite small with the largest difference being 0.15:

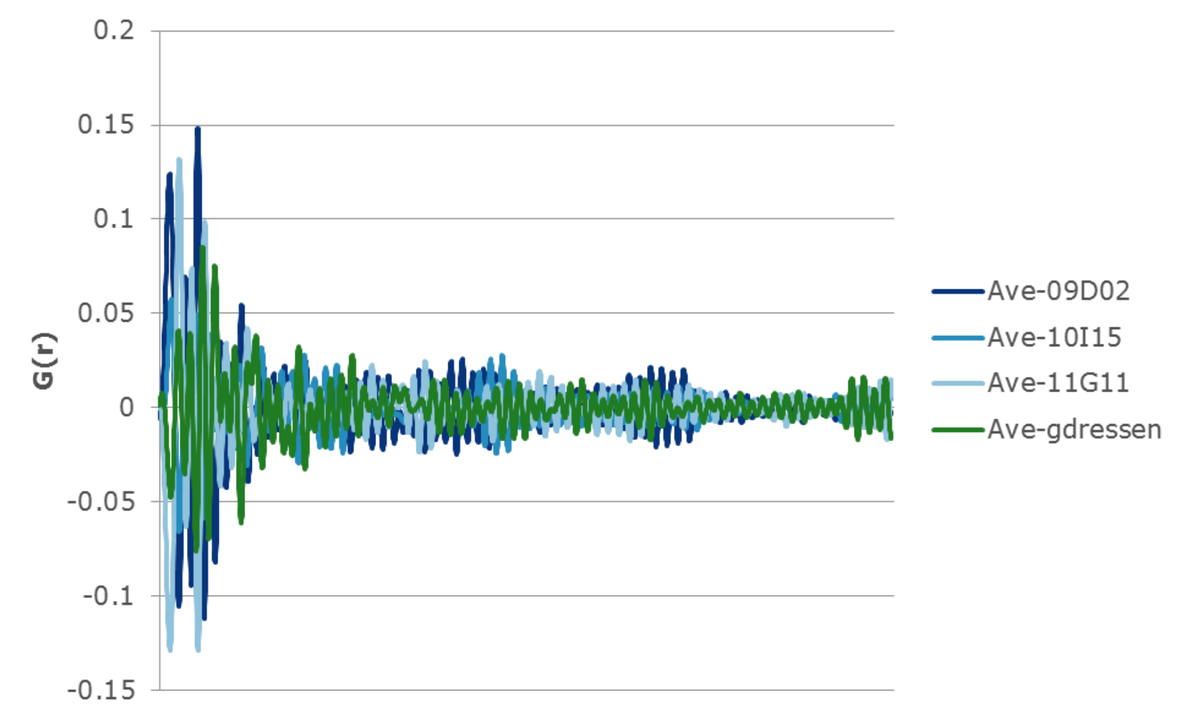
G(r)
Ave-09D02
Ave-10I15
Ave-11G11
Ave-gdressen

An amorphous solid has short-range molecular order but it does not have any long-range molecular order or packing as a crystalline form would. The synchrotron data confirmed that all amorphous SDP formulations prepared using different process are equivalent and show the same structure by PDF synchrotron analyses. Furthermore, samples are equivalent and show no ordering (no crystalline order) beyond 7Å (see Figure below, horizontal axis), so they are true amorphous materials lacking nano crystalline structure.

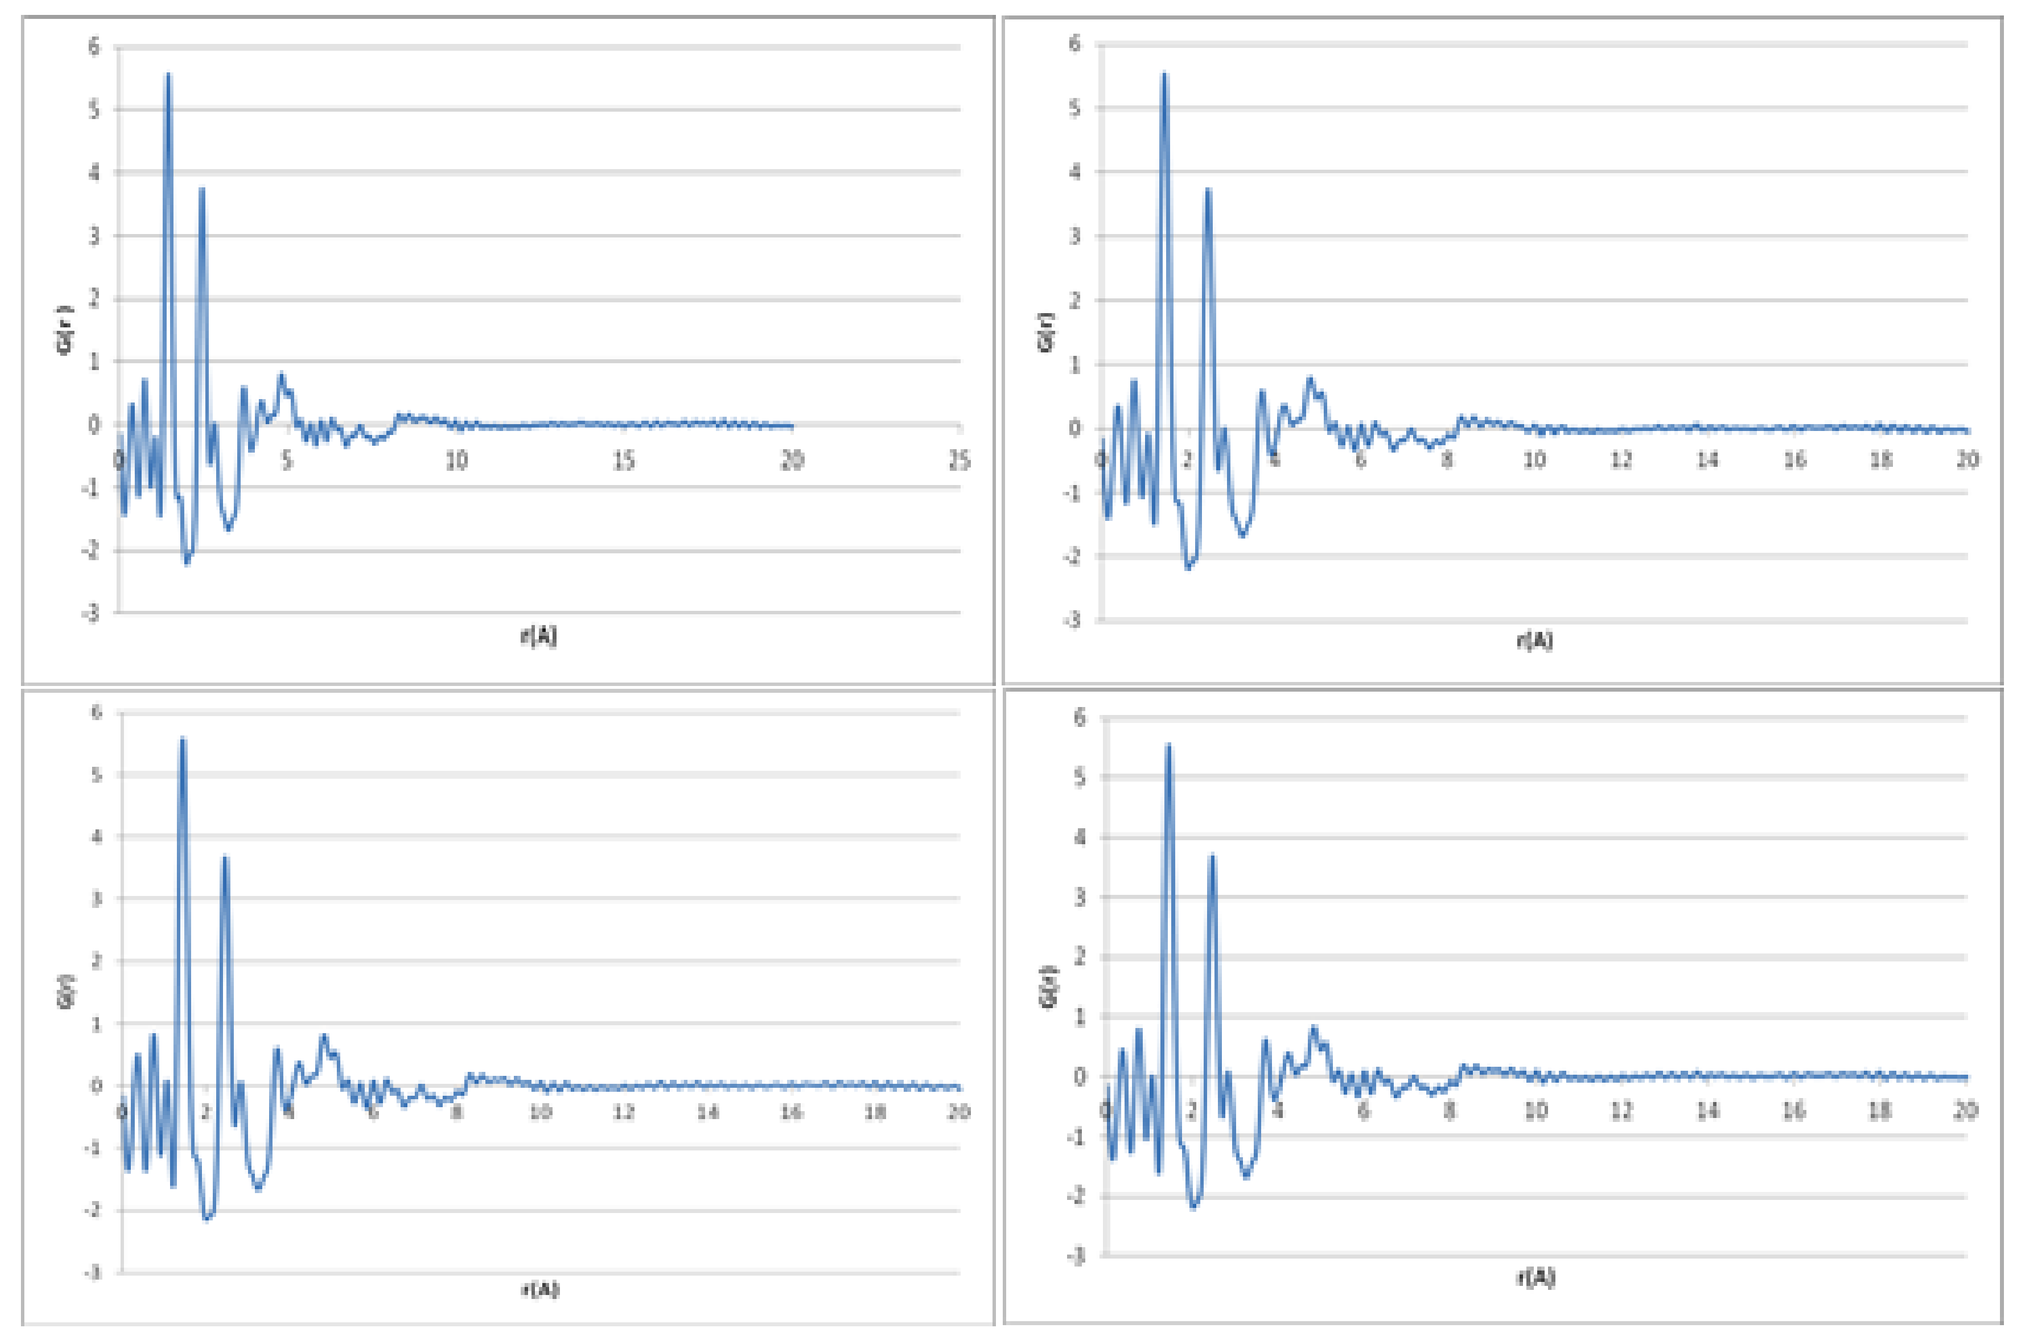

Individual PDF curves for the different analyzed amorphous SDP formulations (top left, clockwise): A, C, B, D

CONCLUSION

PDF analyses was used to confirm the comparability of the amorphous drug prepared by different spray drying processes and gave insight to their degree of molecular order which can be impactful to their physical stability. The PDF traces obtained from synchrotron data, for these SDP formulations, appeared to be more reliable and conclusive than those obtained using laboratory sources. On the other hand, laboratory instrumentation provided for useful insight in a tractable manner and further investigations will be conducted with new x-ray optics and detectors with improved performance for this application .

REFERENCES

1. Boetker JP, Koradia V, Rades T, Rantanen J, Savolainen M; *Pharmaceutics,* 2012 (4), **2012**, 93
2. Dykhne T, Taylor R, Florence A, Billinge SJL; *Pharm. Res*., 28, **2011**, 1041
3. Newman A, Engers D, Bates S, Ivanisevic I, Kelly R C, Zografi G; *J. Pharm. Sciences,* 97 (11), **2008**, 4840.
4. Petkov V; *J. Appl. Cryst* 22, **1989**, 387
5. Qiu X, Thompson J.W., Billinge JL; *J. Appl. Cryst*. 37, **2004**, 678

,

Janssen Research & Development